Modeling of Quality-Adjusted Life-Years (QALYs) Lost Due to Exacerbation of Chronic Obstructive Pulmonary Disease (COPD)

Eva Lydick PhD, Lovelace Clinic Foundation; Leila S. Nelson PhD, Stats 4 U; Herbert T Davis PhD, Lovelace Clinic Foundation; Douglas W Mapel MD MPH, Lovelace Clinic Foundation; Jeno S Marton MD, Pfizer Outcomes Research Group.

Abstract

Aims: To develop estimates of QALYs lost during exacerbations of COPD.

Background: A number of investigators have developed cost-effectiveness models of various therapies for COPD. These models have included QALYs lost due to exacerbations of the disease, as well as costs of treating those exacerbations. However, there are no available data that directly assess QALYs lost during exacerbations of COPD or even quality-of-life changes during an exacerbation. Previous models used QALY weights based on expert opinion with and without reference to published data.

Methods: We modeled QALY decrements for severe and nonsevere exacerbations in patients with severe and mild/moderate COPD based on 1) shape and duration noted by changes in the peak expiratory flow rate over the duration of an exacerbation and 2) the decrement from presentation-to-recovery change in quality-of-life reported for COPD patients seen in an ambulatory clinic. We used a beta-function to model the shape of the exacerbation curve. The duration of the exacerbation in days and the decrement in quality-of-life terms were random variables derived from distributions reported from the two sources mentioned above. The QALY decrement associated with a mild/moderate or severe exacerbation was determined by Monte Carlo simulation. The likelihood of a severe or nonsevere exacerbation was driven by the experience of all patients within Lovelace Health system with a diagnosis of COPD. Utility values for entry level were based on EQ-5D responses from patients with severe and mild/moderate COPD participating in the US Medical Expenditure Panel.

Results: The impact of improved therapy was greatest for patients with mild/moderate COPD, with an overall gain of approximately 4.3 QALYs per 1000 person-year by preventing mild exacerbations and approximately 1.6 QALYs per 1000 person-years in prevention of severe exacerbations. In comparison the QALYs gained by preventing exacerbations among patients with severe COPD were 0.3 and 0.1, respectively.

Conclusions: Our model of QALY decrements due to exacerbations provided a useful and intuitively appealing estimate for the effectiveness of competing therapies for treating patients with COPD. Rather than resorting to expert opinion, effort should be given to modeling the effectiveness measure from available information or, better yet, attempting to measure effectiveness (QALYs) directly.

Background

Four cost-effectiveness analyses for the treatment of chronic obstruct pulmonary disease (COPD) have been used for different ways to estimate the quality-adjusted life-year (QALY) gained from preventing exacerbations of the disease. None of the estimates of the QALY decrement used actual data.

Cost-Effectiveness Analysis	Estimate	Basis of QALY Estimate	Value
Borg et al [1]	Decrement- mild and moderate exacerbation	Expert Panel	mild exacerbation: 5% decrease from baseline state moderate exacerbation: 15% decrease from baseline state
	Decrement – severe extrapolation	Extrapolation from QALY from asthma [2]. Asthmatics asked to assess three marker states using feeling thermometer. Mean for current state = 81; for mild exacerbation, 62; for severe exacerbation, 26.	70% decrease
	Duration	From Seemungal et al [3]	
Oostenbrink et al [4]	Decrement – Nonsevere exacerbation	From Patterson et al [5] -- patients presenting with an acute exacerbation vs end of treatment: EQ-5D changes 0.17	15% decrease from baseline state
	Decrement—Severe exacerbation		50% decrease from baseline state
	Duration	Not stated	Markov cycle was 1 month, so perhaps duration was assumed to be 1 month
Sin et al [6]	Decrement	Oregon Survey	0.32 decrement
	Duration	Model Assumption	Mild: 1 week Moderate: 2 weeks Severe: 4 weeks
Spencer et al [7]	Decrement	Panel of 27 respiratory physicians using EQ-5D	
	Duration	Model Assumption	Logarithmic recovery curve over a period of 90 days with recovery to 0.03 units of baseline

Methods

Our estimation of QALY involved three stages:

•Estimation of the shape of the exacerbation

•Estimation of the extent of the exacerbation at the greatest impairment

•Estimation of the duration of the exacerbation

We also wanted to preserve the variability in the effect of the exacerbation on patients and we wanted to account for both mild exacerbations as well as severe exacerbations.

Shape:

We assumed that the effect of an exacerbation on patients' quality of life (QoL) would tend to follow the pattern of decrease in respiratory function. Seemungal et al have shown that symptoms largely correspond to the decrease in peak expiratory flow rate (PEFR) [3]. Thus we attempted to model the shape based on their reported median PEFRs for COPD patients experiencing exacerbations.

Note peak of exacerbation at day 1

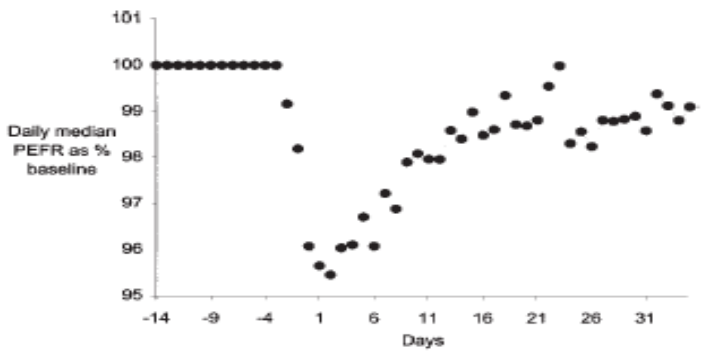
Daily median PEFR as % baseline
Days

Figure 3. Median peak flow expressed as a percentage of baseline peak flow from 14 d before, to 35 d after onset of exacerbation for 504 exacerbations in 91 patients.

We modeled the shape of the exacerbation with a beta function, retaining peak decrement at day 1.

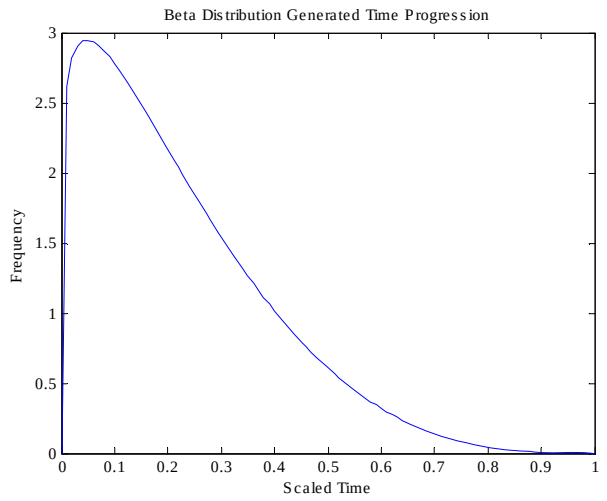
Beta Distribution Generated Time Progression
Frequency
Scaled Time

Methods cont'd

Extent:

Patterson et al [5] provide a measure of 'responsiveness' – or change for people that show up at a physician's office with an exacerbation of COPD. From their tables, we extrapolated the mean change on EQ-5D for those who felt they were 'a lot better' after treatment with antibiotics. We therefore assumed an equivalent change for usual health to peak of exacerbation. Since these patients were all treated as outpatients, we considered them to have mild exacerbations.

Andersson [2] reported a decrement of 0.55 in patients with a severe asthma exacerbation, so we assumed the same value for severe exacerbations of COPD, based on clinical judgment that the two groups of patients were similarly ill. Utility values usually have a large variance, but in order to not span the entire spectrum from 0 to 1, we set the standard deviation at 1/2 the estimate of the mean decrement.

Thus we simulated extent

• Using a normal distribution

•Mild or moderate exacerbation

$\mu = 15\%, \sigma = 7.5\%$

•Severe exacerbation

$\mu = 55\%, \sigma = 27.7\%$

Duration:

From Seemungal et al [3] median duration for mild exacerbations was 6 days and interquartile range (IQR) was [1,14] and for severe exacerbations the IQR was [28,42] days.

• We modeled duration using a gamma distribution

•Mild or moderate exacerbation

K=.715, λ=14.23

•Severe exacerbations

K=11.37, λ==3.13

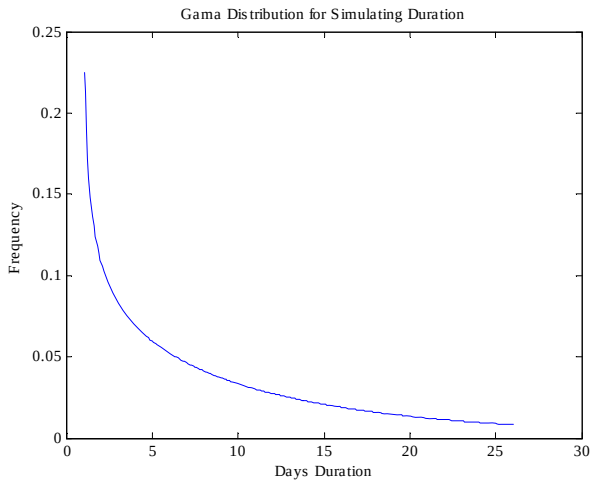
Gama Distribution for Simulating Duration
Frequency
Days Duration

This project is sponsored by a grant from GlaxoSmithKline

Results

The QALY decrement for a mild or severe exacerbation was estimated by Monte Carlo simulation from distributions for duration and extent. The QALY was the integral of the decrement expressed as:

$$QALY = \int_0^{duration} f(x)\,dx = \frac{extent\ *\ duration}{2.9}$$

The QALY thus estimated was incorporated into a cost-effectiveness analysis that generated costs for each patients (drug costs and healthcare utilization costs for exacerbation, if predicted) and QALY decrement (if exacerbation occurred). The difference between the total QALY decrements for two different treatments makes up the denominator of the cost-effectiveness ratio. The numerator was the difference in cost between the two treatment groups. The figure below represents results of 1000 runs using the patient experience of approximately 2400 patients with COPD in the Lovelace Health System database.

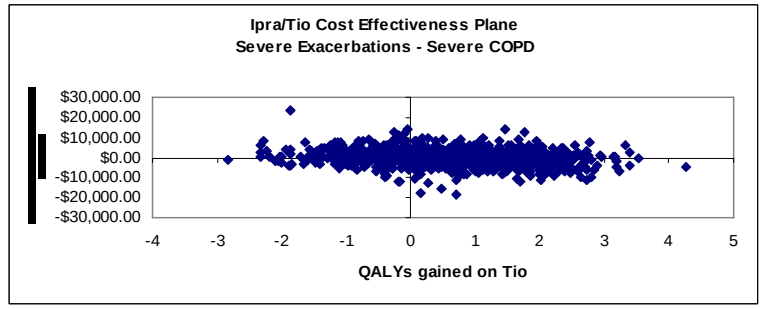
Ipra/Tio Cost Effectiveness Plane
Severe Exacerbations - Severe COPD
QALYs gained on Tio

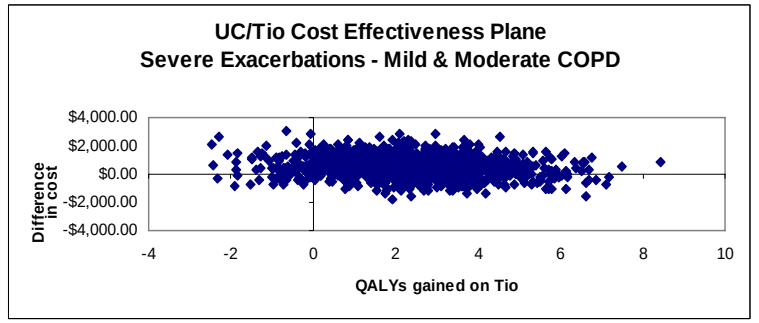
UC/Tio Cost Effectiveness Plane
Severe Exacerbations - Mild & Moderate COPD
Difference in cost
QALYs gained on Tio

Conclusions

Our model of QALY decrements due to exacerbations provided a useful and intuitively appealing estimate for the effectiveness of competing therapies for treating patients with COPD. However, given the number of cost-effectiveness analyses of therapies to prevent exacerbations in patients with COPD, the question is why effort has not been to measuring QALYs directly.

References

1. Borg S, Ericsson A, Wedzicha J, Gulsvik A, Lundback B, Donaldson GC, Sullivan SD. A computer simulation model of the natural history and economic impact of chronic obstructive pulmonary disease. Value Health 2004; 7:153-167.
2. Andersson F, Borg S, Stahl E. The impact of exacerbations on the asthmatic patient's preference scores. J Asthma 2003; 40:615-630.
3. Seemungal TA, Donaldson GC, Bhowmik A et al. Time course and recovery of exacerbations in patients with chronic obstructive respiratory disease. Am J Respir Crit Care Med 2000; 161:1608-1613.
4. Oostenbrink JB, Rutten-van Molken MPMH, Monz BU, FitzGerald JM. Probabilistic Markov model to assess the cost-effectiveness of bronchodilator therapy in COPD patients in different countries. Value Health 2005; 8: 32-46.
5. Paterson C, Langan CE, McKaig GA et al. Assessing patient outcomes in acute exacerbations of chronic bronchitis: the measure your medical outcome profile (MYMOP), medical outcomes study 6-item general health survey (MOS-6A) and Euro-Qol (EQ-5D). Qual Life Res 2000; 9:521-527.
6. Sin DD, Golmohammadi K, Jacobs P. Cost-effectiveness of inhaled corticosteroids for chronic obstructive pulmonary disease according to disease severity. Am J Med 2004; 116:325-331.
7. Spencer S, Jones PW. Time course of recovery of health status following an infective exacerbation of chronic bronchitis. Thorax 2003; 58:589-593.